Positive ad hoc results from neoadjuvant Foxy-5 treatment of colon cancer patients in the ongoing phase 2 NeoFox study

Ramon Salazar Soler[1], C. Santos Vivas[2], I. Macias[3], A. Yubero Esteban[4], A. Bassam[5], J. Gallego Plazas[6], Y. Al-Farhat[7], A. Cervantes[8], P. García Alfonso[9], D. Paez[10], A. Ruiz-Casado[11], C. Lopez[12], P. F. Árkosy[13], A. Furka[14], N. Rodriguez-Salas[15], F. J. Ramos Pascual[16], A. Cubillo Gracian[17], E. Hitre[18], R. Ferreiro Monteagudo[19], M.A. Salud Salvia[20]

Scientific rationale

High tumour-expression of WNT5A is associated with better overall survival in colon cancer.
A WNT5A-mimicking peptide, Foxy-5, was developed and shown to effectively reduce cancer cell invasion and metastasis in pre-clinical models.

Methodology

A randomised, controlled, and open multicenter phase 2 clinical study was conducted in patients with stage II/III colon cancer. Patients received three doses (1.8 mg/kg) per week of neoadjuvant Foxy-5 treatment for at least 3 weeks, followed by up to 10 weeks of adjuvant treatment. Primary efficacy evaluations were based on ctDNA relapse (analyses still ongoing). An ad hoc analysis was performed on surgically removed material from 110 patients at the time indicated by the red arrow (figure 1).

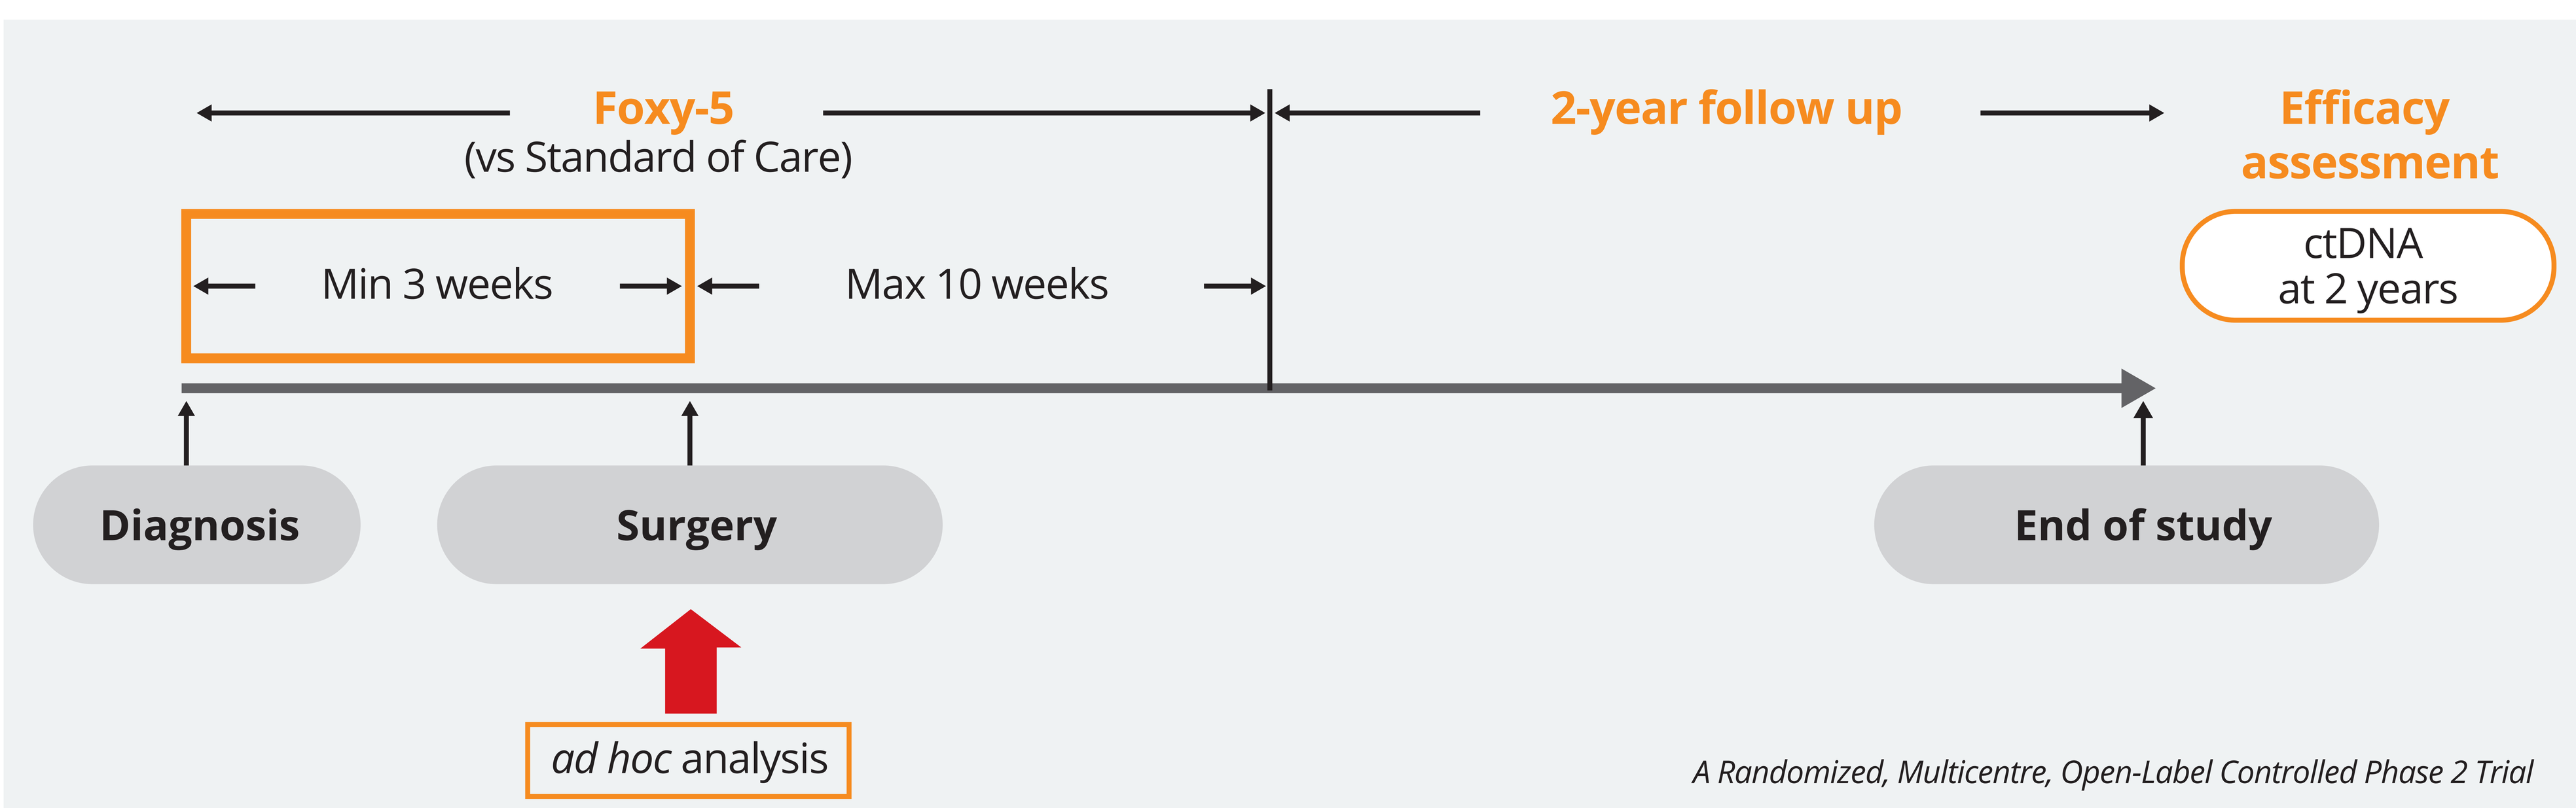
Foxy-5
(vs Standard of Care)
2-year follow up
Efficacy assessment
Min 3 weeks
Max 10 weeks
ctDNA
at 2 years
Diagnosis
Surgery
End of study
ad hoc analysis
A Randomized, Multicentre, Open-Label Controlled Phase 2 Trial

▲ **Figure 1** Original study design

Safety

Based on 127 patients (64 patients in the Foxy-5 arm, treatment period completed), there were 27 Foxy-5 related adverse events, 26 of which were Grade 1 and one Grade 2 (table 1). Foxy-5 was considered safe and well-tolerated.

▼ **Table 1** Foxy-5 safety profile

	Any AE / related	Serious / related
Foxy-5 arm	165 / 27	6 / 0
Control arm	41 / 0	1 / 0

	Grade 1	Grade 2	Grade 3	Grade 4
Foxy-5 related AE	26	1	-	-
Control any AE	28	9	4	-

Results

Pathological examination suggested an effect of Foxy-5 on the local spread of the primary tumours within 3-4 weeks of treatment:

- Fewer patients had perineural invasion (p=0.0088, figure 2)
- Fewer patients had venous invasion (p=0.0097, figure 3)

In addition, Foxy-5 was associated with TN(M) downstaging (p=0.012, figure 4).

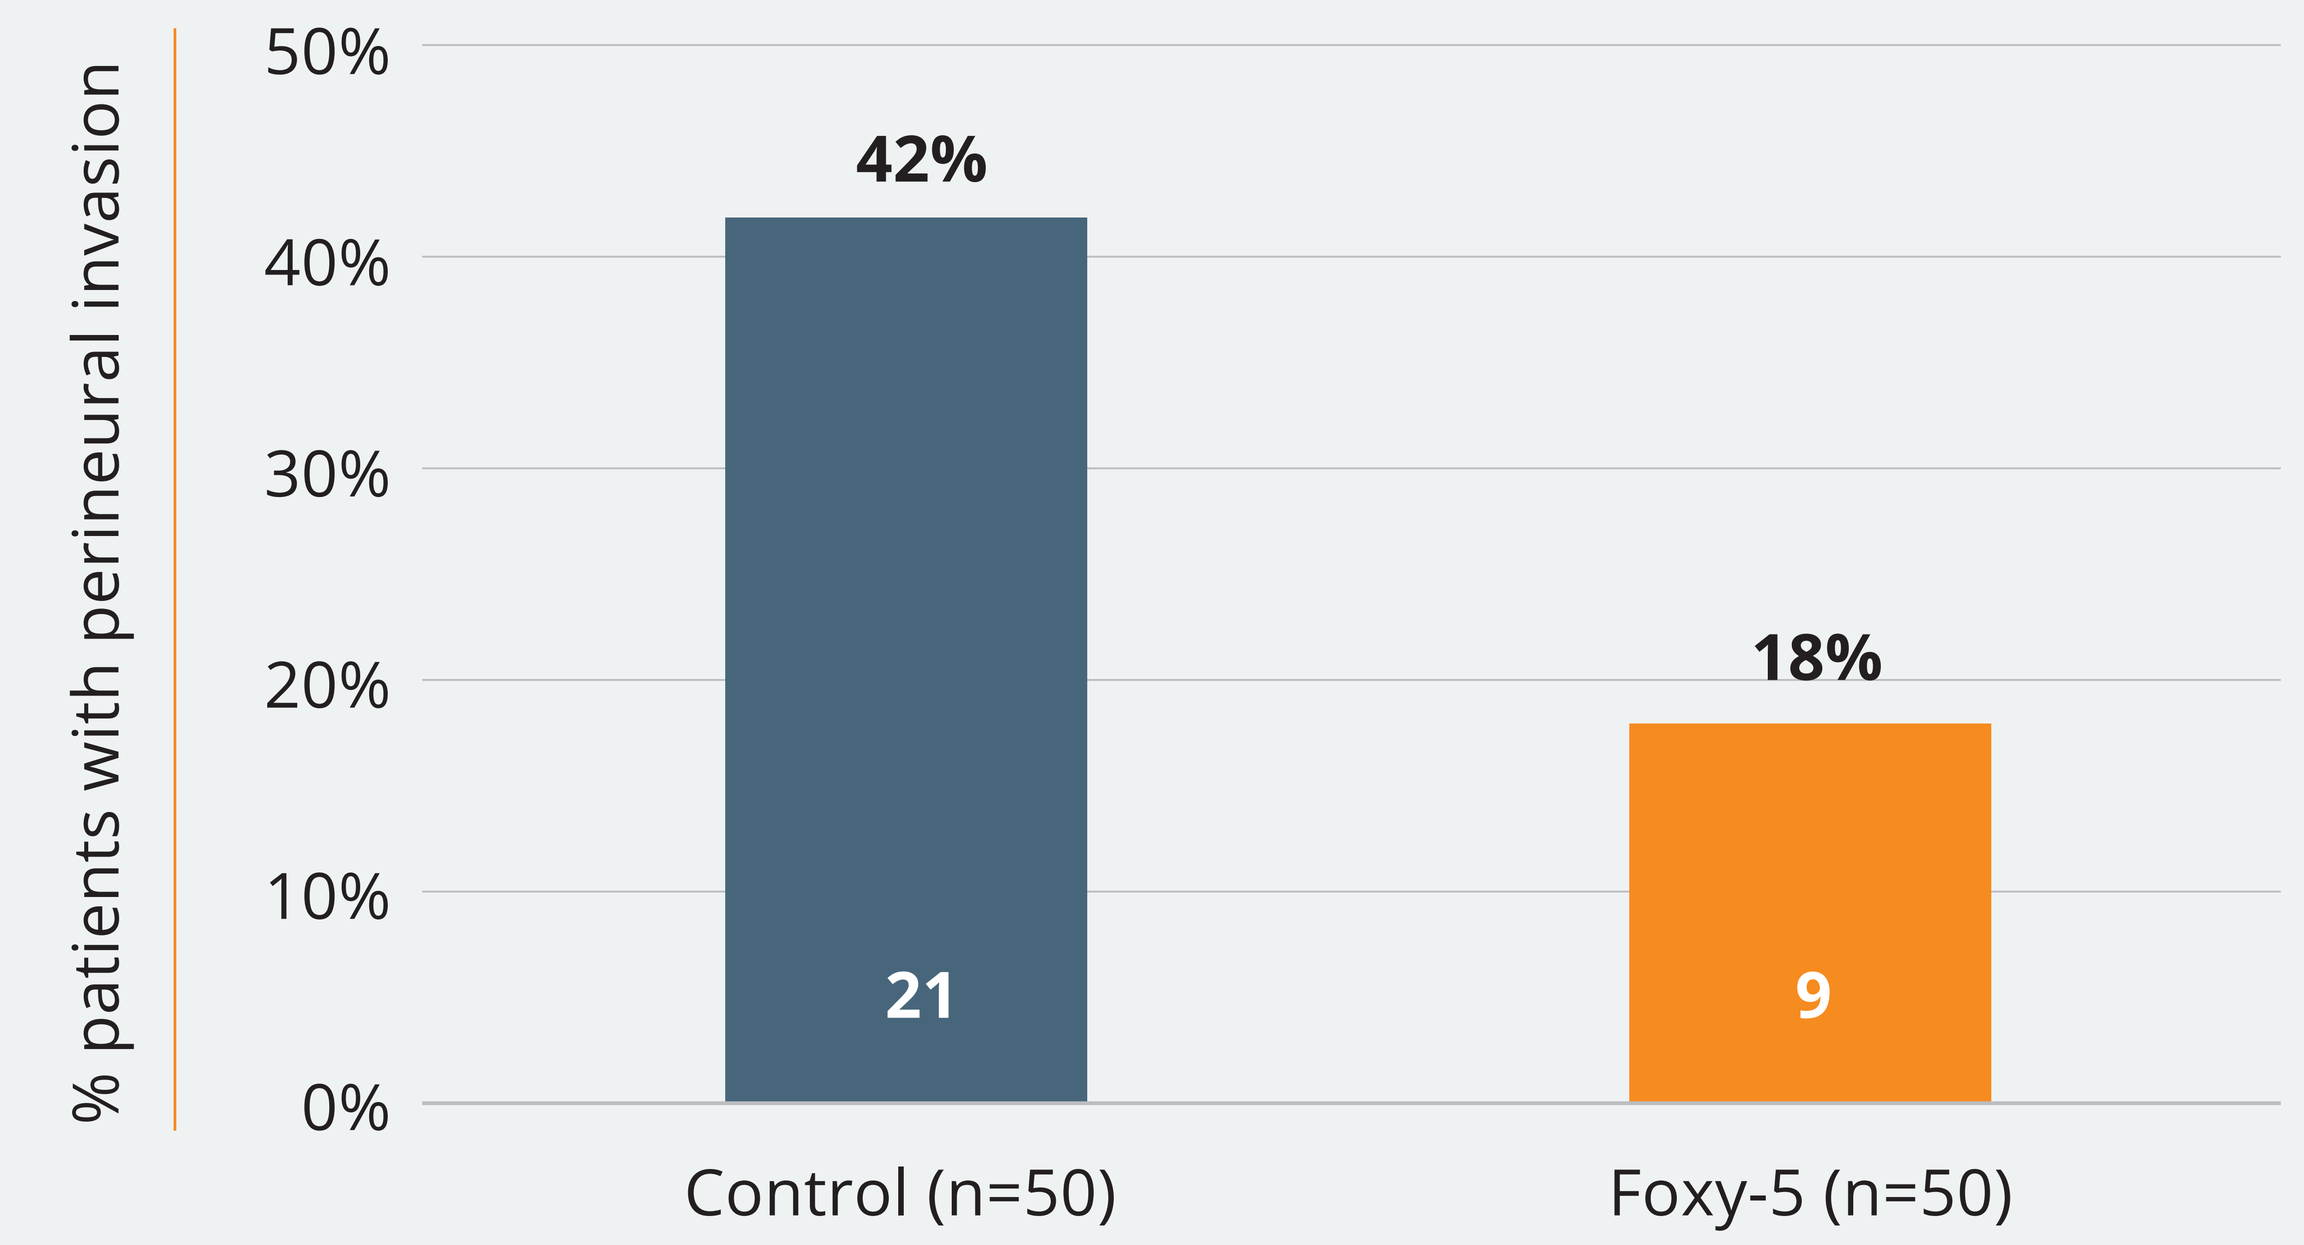
% patients with perineural invasion
50%
40%
30%
20%
10%
0%
42%
21
18%
9
Control (n=50)
Foxy-5 (n=50)

▲ **Figure 2** Histopathological assessment of perineural invasion after 3-4 weeks of treatment with Foxy-5 (n=100).

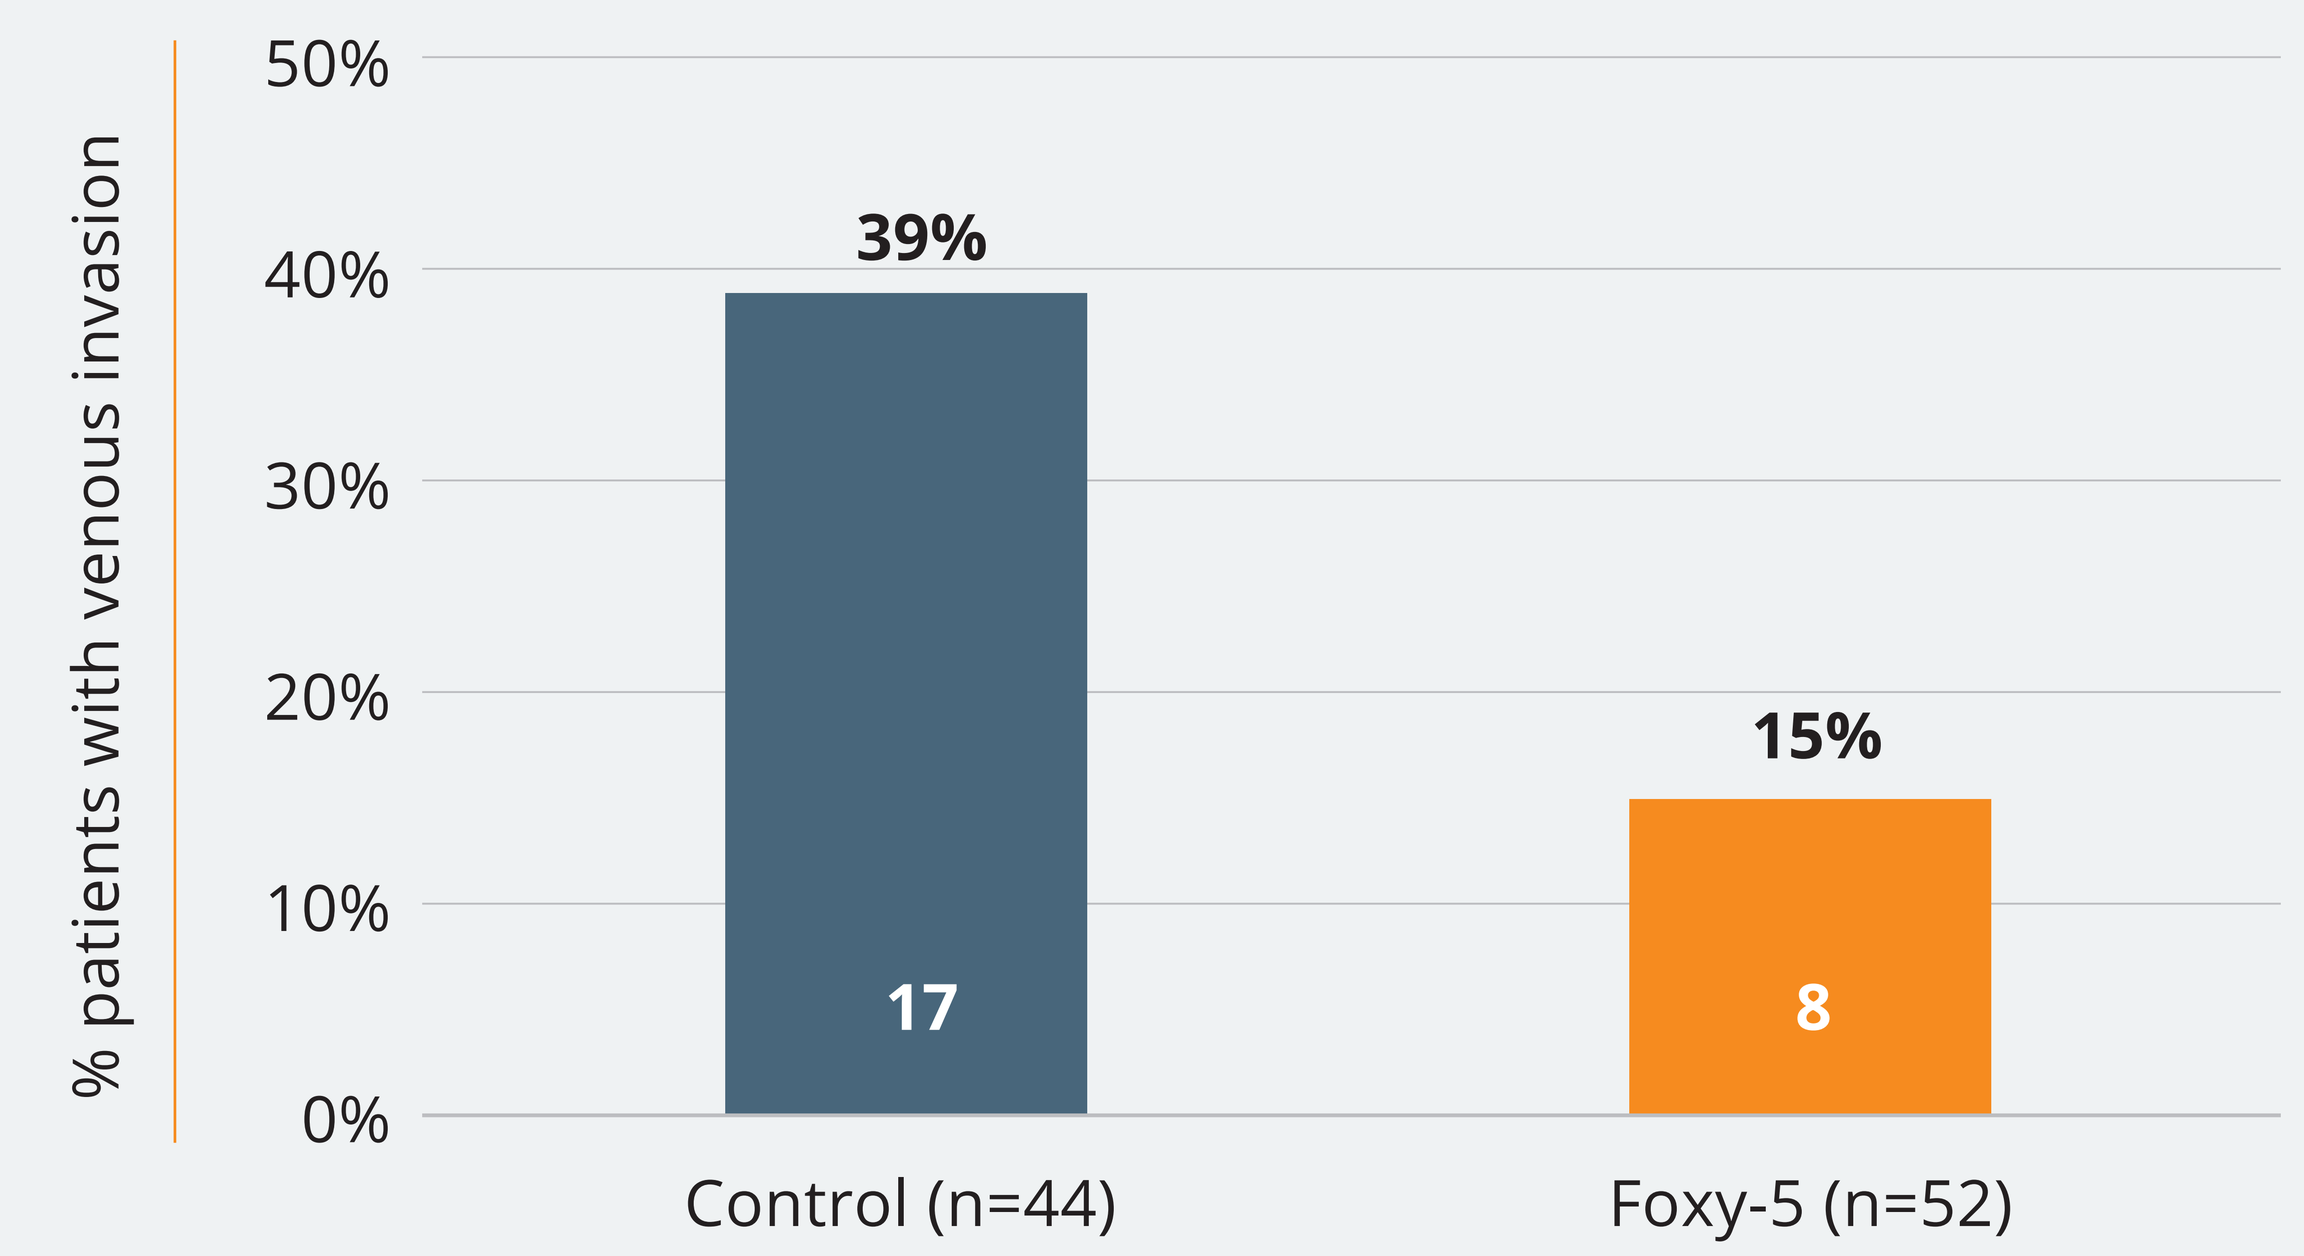
% patients with venous invasion
50%
40%
30%
20%
10%
0%
39%
17
15%
8
Control (n=44)
Foxy-5 (n=52)

▲ **Figure 3** Histopathological assessment of venous invasion after 3-4 weeks of treatment with Foxy-5 (n=96).

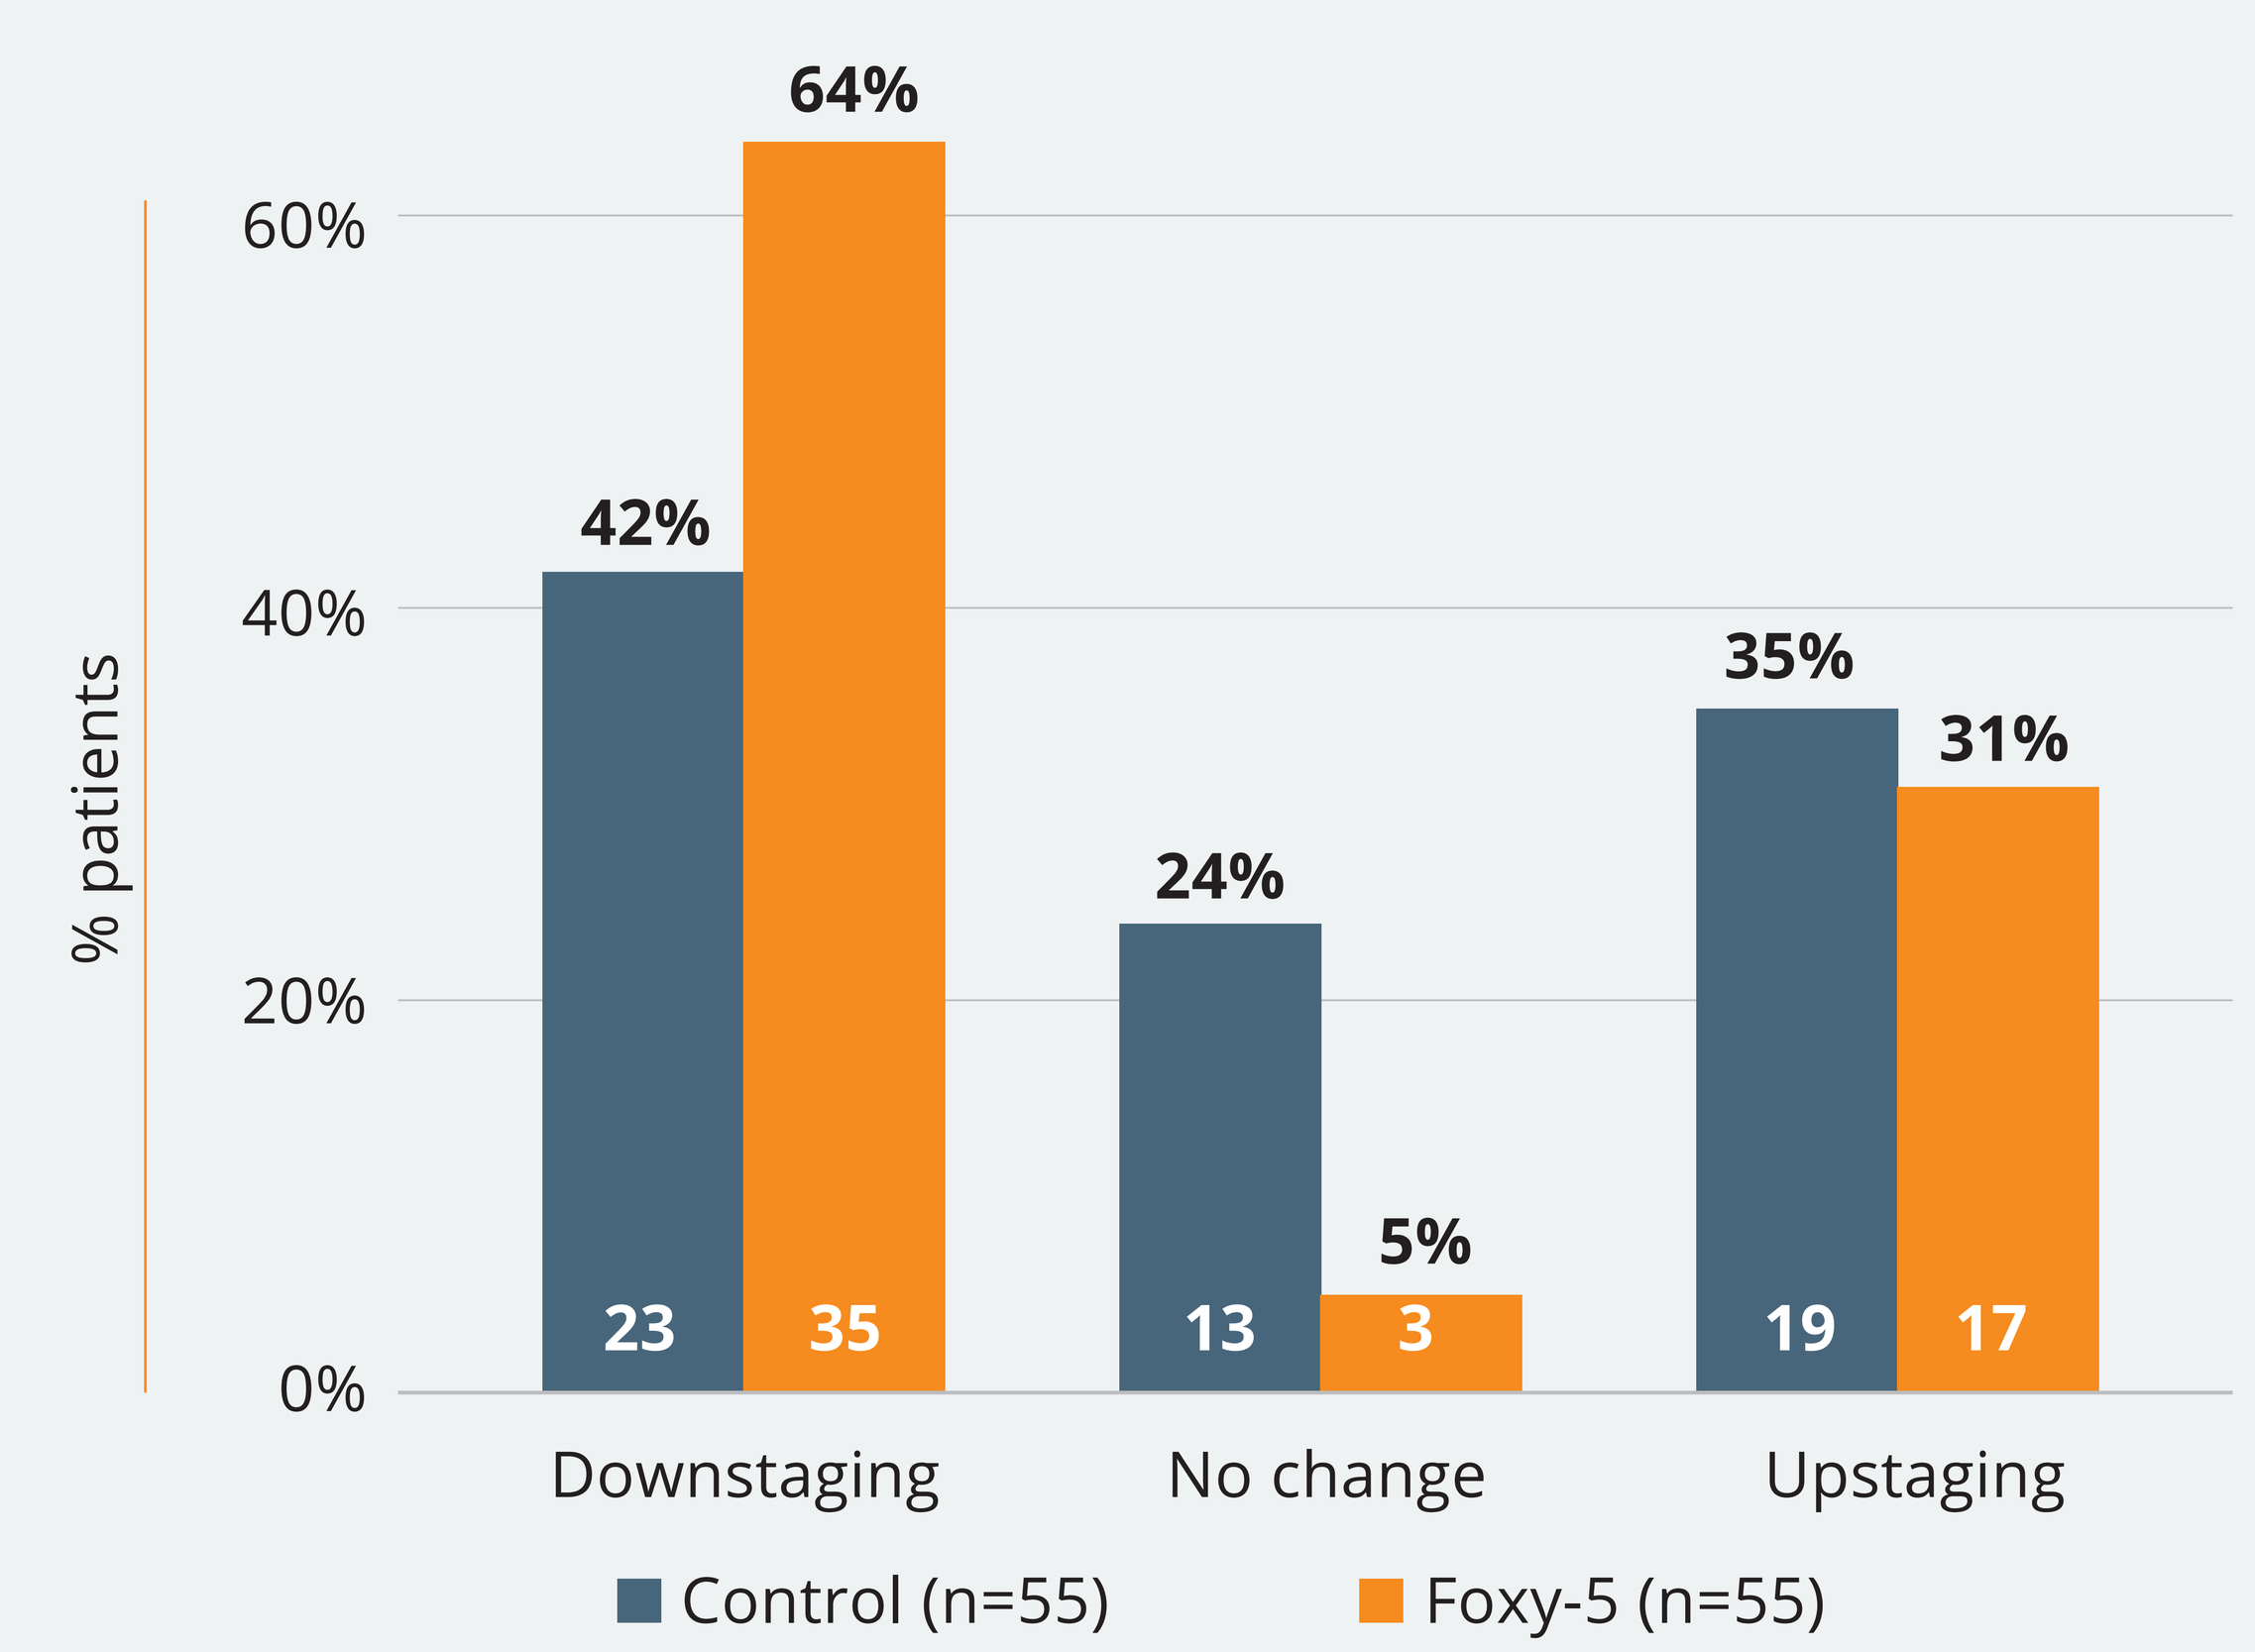
% patients
60%
40%
20%
0%
42%
64%
23
35
24%
5%
13
3
35%
31%
19
17
Downstaging
No change
Upstaging
Control (n=55)
Foxy-5 (n=55)

▲ **Figure 4** TN(M) staging (M not analysed since metastatic disease was an exclusion criteria) was performed by CT at study start, and by histopathological assessment using material from the surgical resection (n=110).

Discussion

The significance of histologic findings in a neo-adjuvant setting is supported by the FOxTROT trial. Six weeks of preoperative FOLFOX treatment produced substantial reductions in T stage, N stage, and EMVI, resulting in better 2-year disease control. Table 2 illustrates an indirect comparison between preoperative FOLFOX and Foxy-5 treatment: fewer patients had venous invasion after 3 weeks of Foxy-5 treatment compared to after 6 weeks of FOLFOX treatment.

▼ **Table 2** Local pathology comparison: FOxTROT vs NeoFox

	FOxTROT		NeoFox	
	Neoadj. chemo (n=684)	Straight to surgery (n=347)	Foxy-5 (n=56)	Control (n=55)
EMVI/venous invasion	32.3%	45.0%	15.4% (n=52)	38.6% (n=44)
pT0	4.1%	0%	0%	0%
pT1/T2	11.7%	5.7%	12.5%	5.5%
pT3	63.6%	63.8%	58.9%	69.1%
pT4	20.7%	30.5%	28.6%	25.5%

Morton, Dion et al., Journal of clinical oncology vol. 41,8 (2023): 1541-1552
Data on file, NeoFox

Implication

The indications of rapid effects of preoperative Foxy-5 on local invasion and TN(M) downstaging in patients with stage II/III colon cancer led to a decision to amend the study to focus solely on short-term neoadjuvant treatment. Also, given the excellent safety profile of Foxy-5, a dose-escalation was performed, which indicated that 8 mg/kg of Foxy-5 given 5 times per week was safe and well-tolerated.

New study design

The new study evaluates the effects of at least 3 weeks of preoperative Foxy-5 treatment on tumour growth and local spread in patients with stage III colon cancer. Patients (n=80) are randomly assigned to receive either Foxy-5 or standard of care. CT assessments are performed at the study start and pre-surgery, and resected material is analysed.

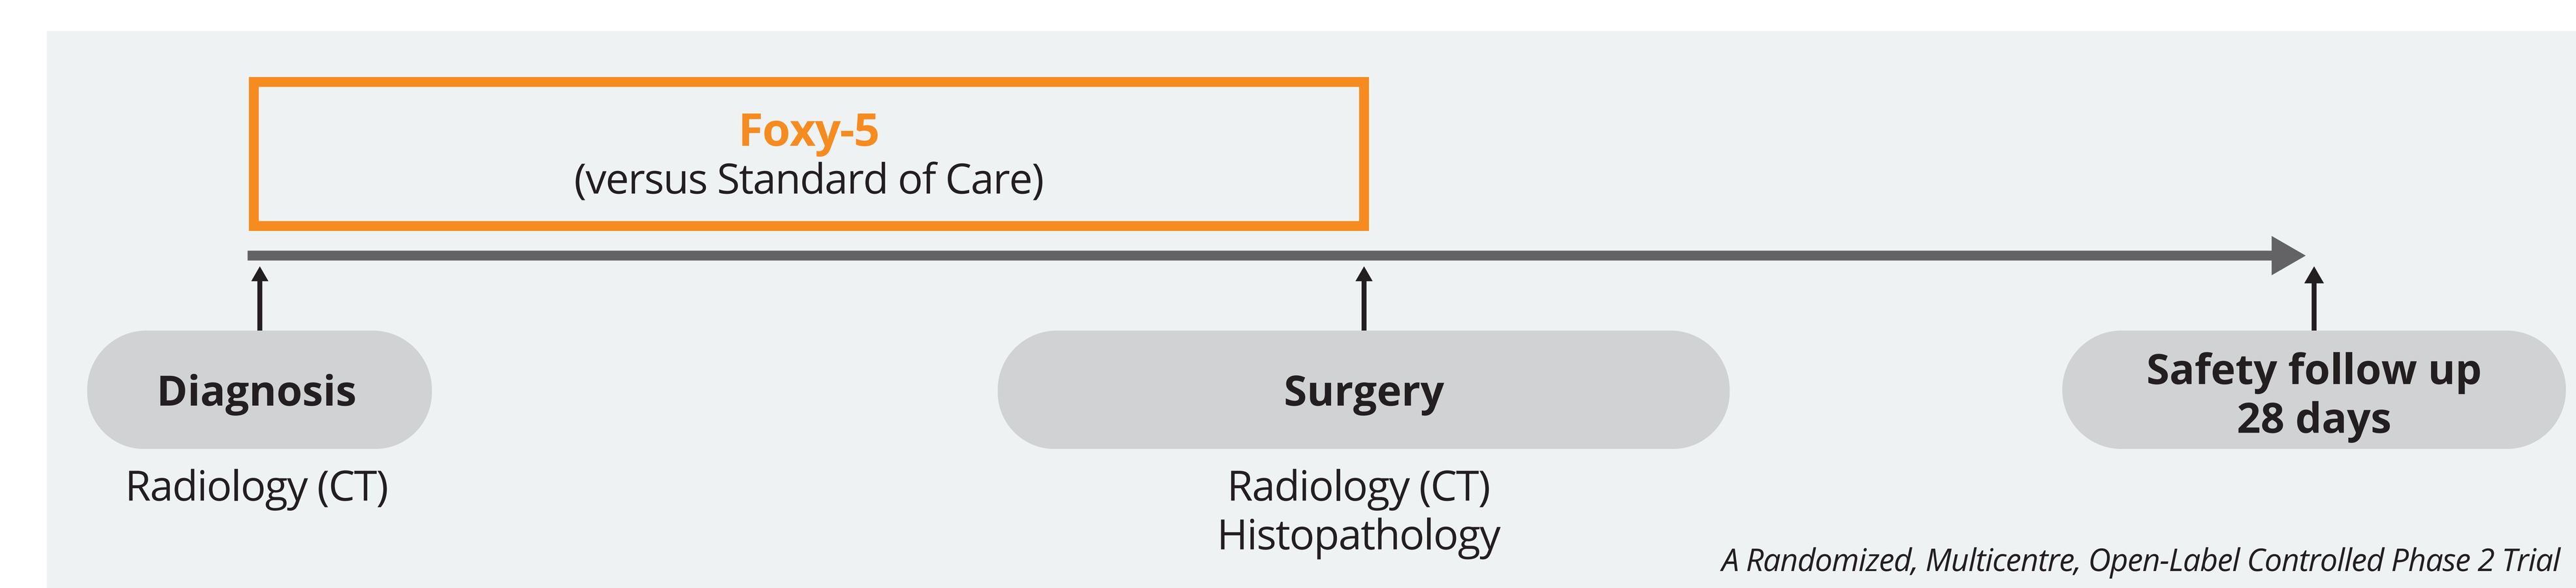
Foxy-5
(versus Standard of Care)
Diagnosis
Radiology (CT)
Surgery
Radiology (CT)
Histopathology
Safety follow up
28 days
A Randomized, Multicentre, Open-Label Controlled Phase 2 Trial

▲ **Figure 5** New study design

Conclusion

Preoperative Foxy-5 treatment was associated with an excellent safety profile, reduced perineural and venous invasion, and TN(M) downstaging in patients with stage II/III colon cancer. Foxy-5 is being evaluated as a potential neoadjuvant treatment for stage III colon cancer.

Author affiliations.
1. Institut Catala d'Oncologia - Hospital Duran i Reynals (ICO L'Hospitalet), Hospitalet De Llobregat, Spain; 2. Institut Català d'Oncologia l'Hospitalet (Hospital Duran i Reynals), L'Hospitalet De Llobregat, Spain; 3. Parc Taulí Hospital Universitari, Barcelona, Spain; 4. Hospital Clinico Universitario Lozano Blesa, Zaragoza,Spain; 5. Pandy Kalman Hospital, Gyula, Hungary; 6. Hospital General Universitario de Elche, Elche, Spain; 7. Tolna County Teaching Hospital, Szekszárd, Hungary; 8. Hospital Clinico Universitario de Valencia, Valencia, Spain; 9. Hospital General Universitario Gregorio Maranon, Madrid, Spain; 10. Hospital de la Santa Creu i Sant Pau, Barcelona, Spain; 11. University Hospital Puerta de Hierro Majadahonda, Majadahonda, Spain; 12. HUMV - Hospital Universitario Marques de Valdecilla, Santander, Spain; 13. Debrecen University Clinical Center, Debrecen, Hungary; 14. Debrecen University Clinical Center, Debrecen, Hungary; 15. Hospital Universitario La Paz, Madrid, Spain; 16. Hospitals Santa Tecla, Tarragona, Spain, 17. Hospital HM - Sanchinarro, Madrid, Spain; 18. National Institute of Oncology, Budapest, Hungary; 19. Hospital Universitario Ramon y Cajal, Madrid, Spain; 20. Hospital Universitario Arnau de Vilanova, Lleida, Spain

ESMO GI Annual Meeting 2024

FPN: Poster 9P. Study sponsored by WntResearch AB. ClinicalTrials.gov: NCT03883802.
Contact: ramonsalazar@iconcologia.net
Ramon Salazar Soler has the following conflicts of interest to declare: WntResearch, Esteve, GSK, Servier, SACE Medhealth, Sanofi, TTD
Copies of this poster obtained through QR and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

wntresearch.com